3931

The Aldehyde Trap NS2 Mitigates Dense Haze in a Rabbit Model of Photorefractive Keratectomy

Gibson, Daniel J.[1]; Mandell, Kenneth J.[2]; Young, Scott L.[2]; Brady, Todd C.[2]
[1]University of Florida, Gainesville, FL, United States. [2]Aldeyra Therapeutics, Lexington, MA, United States

DISCLOSURES: Gibson, DJ (F), Mandell, KJ (C), Young SL (E) and Brady TC (E)

Purpose

We sought to test whether a novel anti-inflammatory drug can reduce corneal haze in a rabbit model of photorefractive keratectomy (PRK) when topically applied prior to surgery and in the 5 days following surgery.

METHODS

All of the animals used herein were treated in a manner consistent with the ARVO Statement for the Use of Animals in Ophthalmic and Vision Research.

Treatment: Two groups with 9 NZW rabbits each (18 total animals, 18 eyes per group) received bi-lateral eye drops of either NS2 or the vehicle for 3 days prior to surgery and then for 5 days after, 4 times daily during waking hours.

Surgery: The rabbits were both generally and topically anesthetized prior to surgery. The cornea's central thickness was obtained by ultrasonic pachymetry (n = 5 per cornea) and an image of the cornea was taken by a dSLR and macro lens. A trans-epithelial excimer laser wound with a 6.0 mm diameter by 150 µm deep was created on each cornea. The residual corneal thickness was immediately obtained again by ultrasonic pachymetry. Next, the wound area was recorded by fluorescein instillation and fluorescent macrophotography. Post-surgical analgesia was provided by daily oral meloxicam until wound closure. The eyes' irritation, corneal thicknesses, and wound sizes were measured the next day.

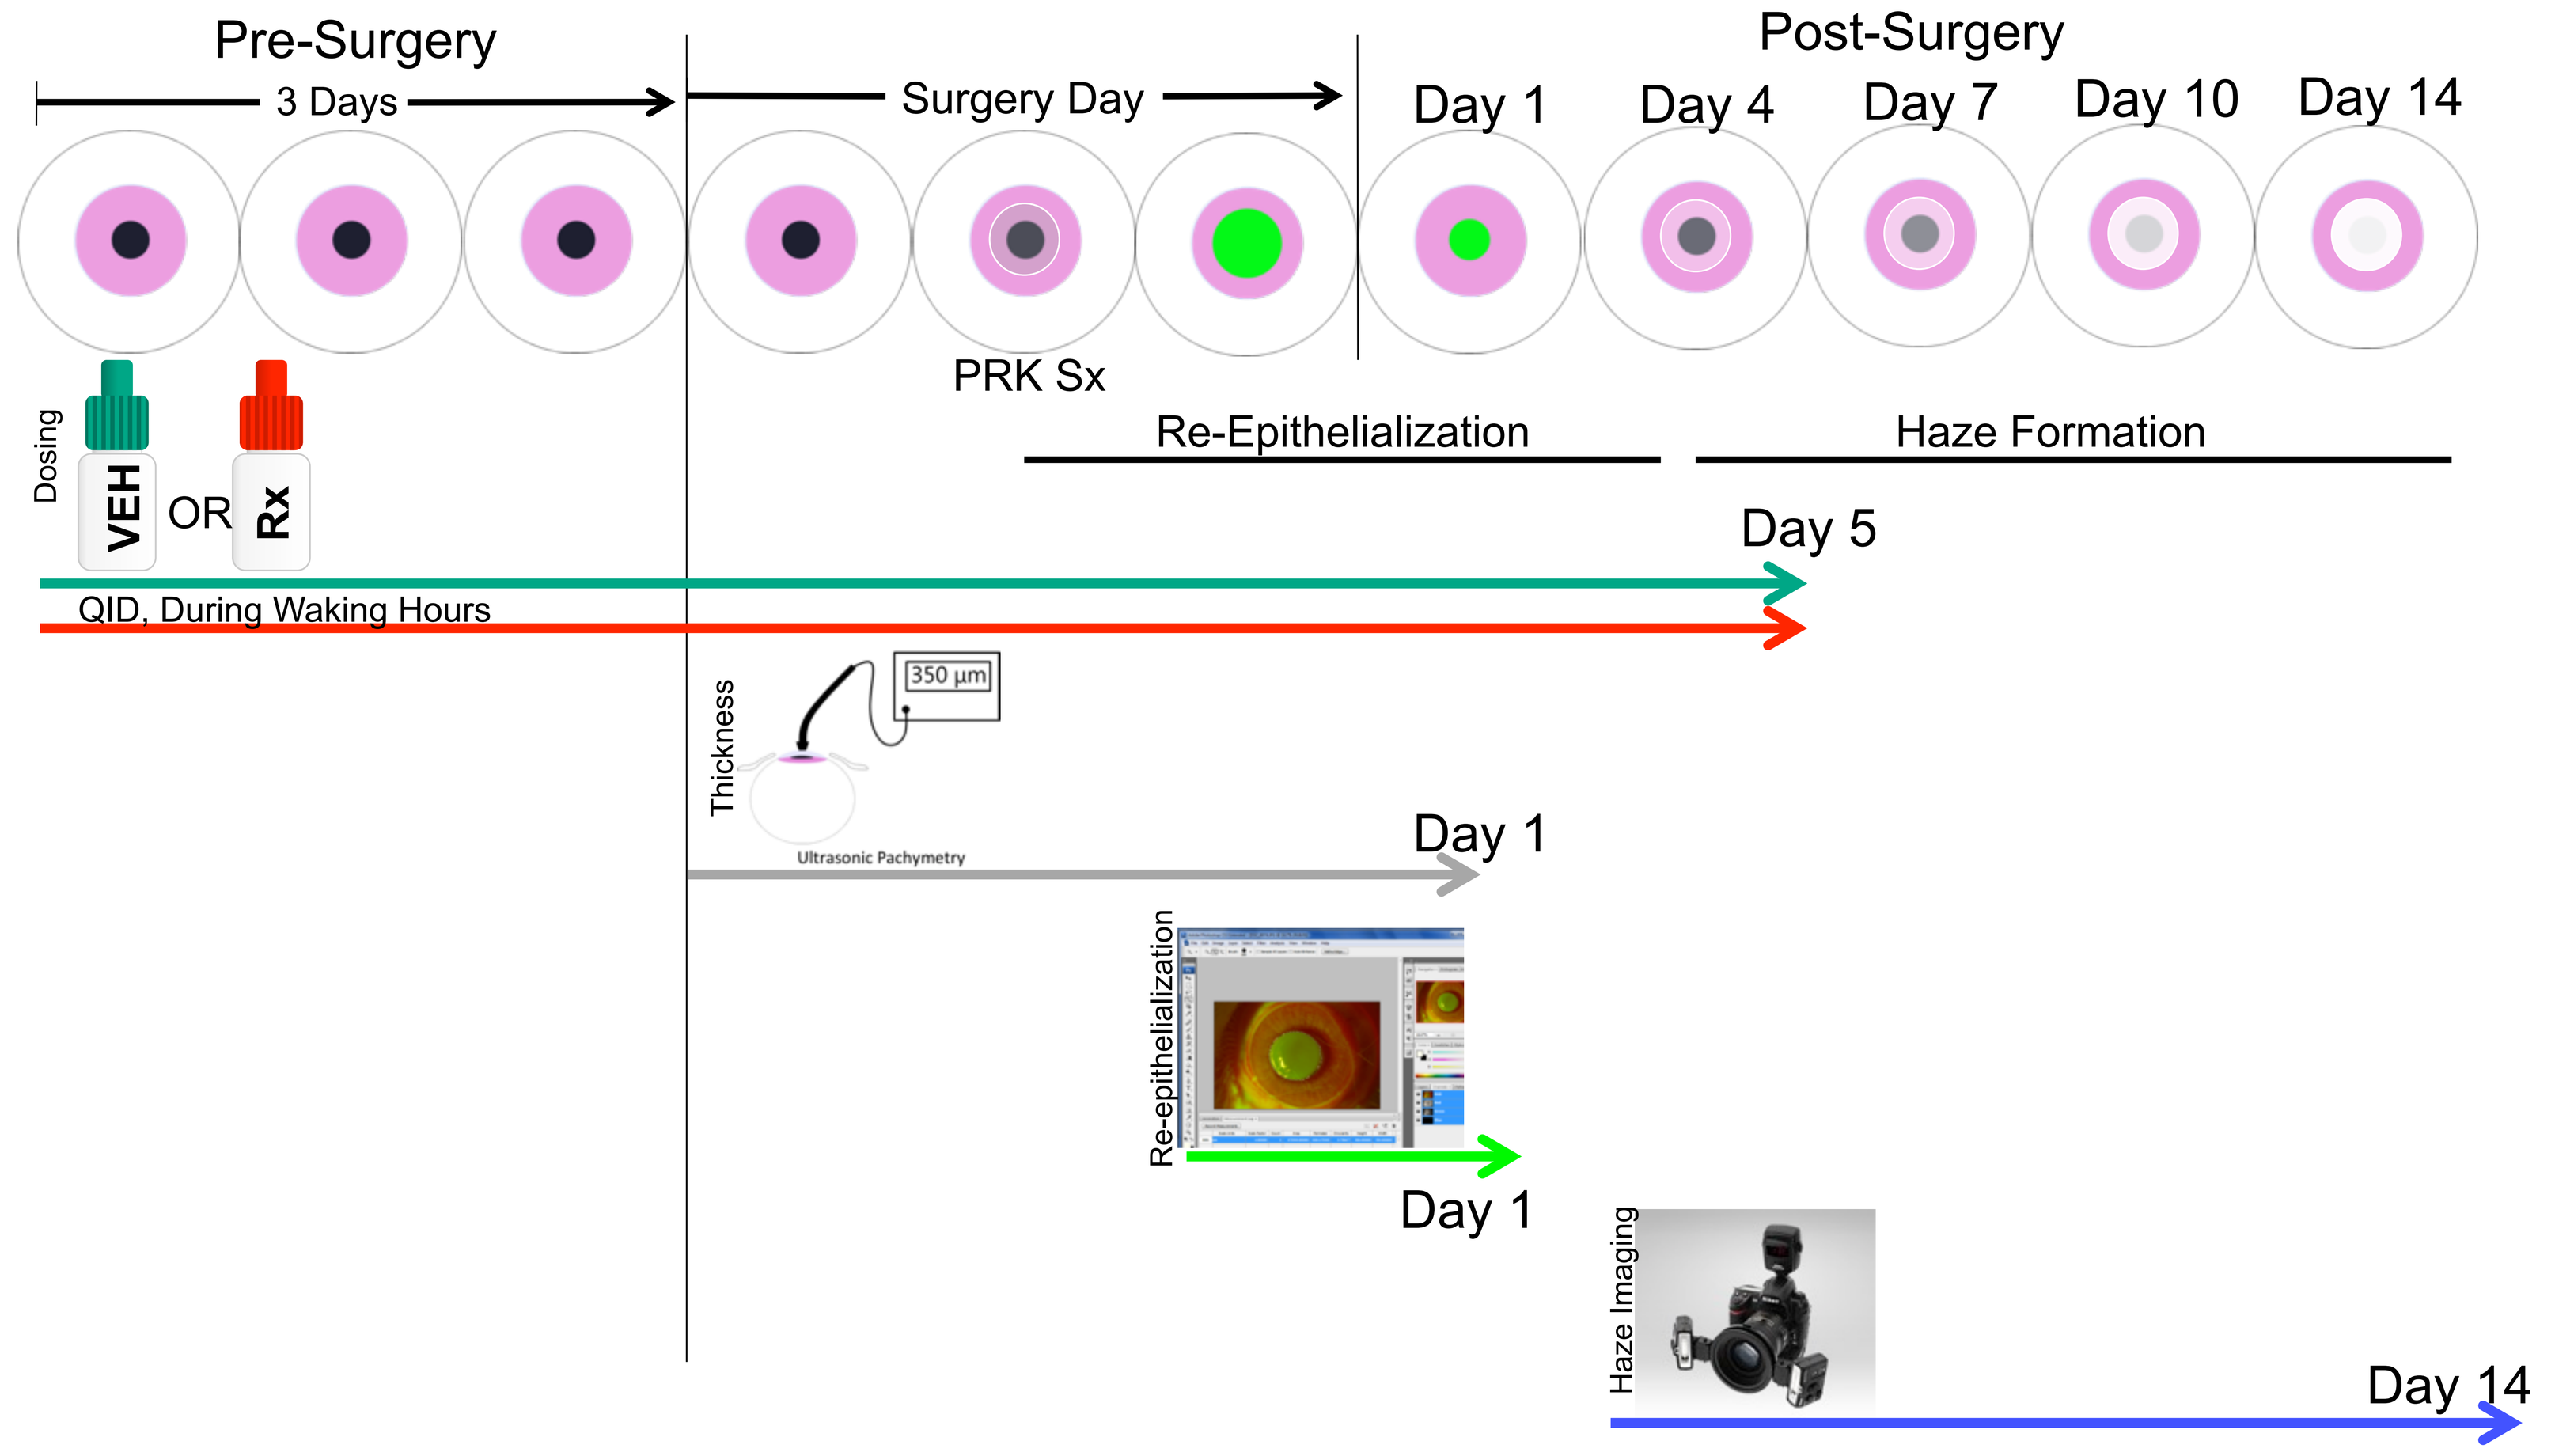
Pre-Surgery
3 Days
Surgery Day
Post-Surgery
Day 1
Day 4
Day 7
Day 10
Day 14
PRK Sx
Re-Epithelialization
Haze Formation
Dosing
VEH
OR
Rx
Day 5
QID, During Waking Hours
Thickness
350 µm
Ultrasonic Pachymetry
Day 1
Re-epithelialization
Day 1
Haze Imaging
Day 14

Figure 1. A schematic representation of the experiment, the timing, and the data collected.

Haze Measurements: Digital macro images of the haze taken at days 4, 7, 10, & 14 post surgery were analyzed. First, the images were converted to greyscale utilizing only the data in the blue channel. A consistent circular region surrounding the wound was taken and the histogram was used to compare the retinal and haze peaks. The difference between these two "modes" represented the magnitude of the most abundant haze intensity present, by comparison to the retina.

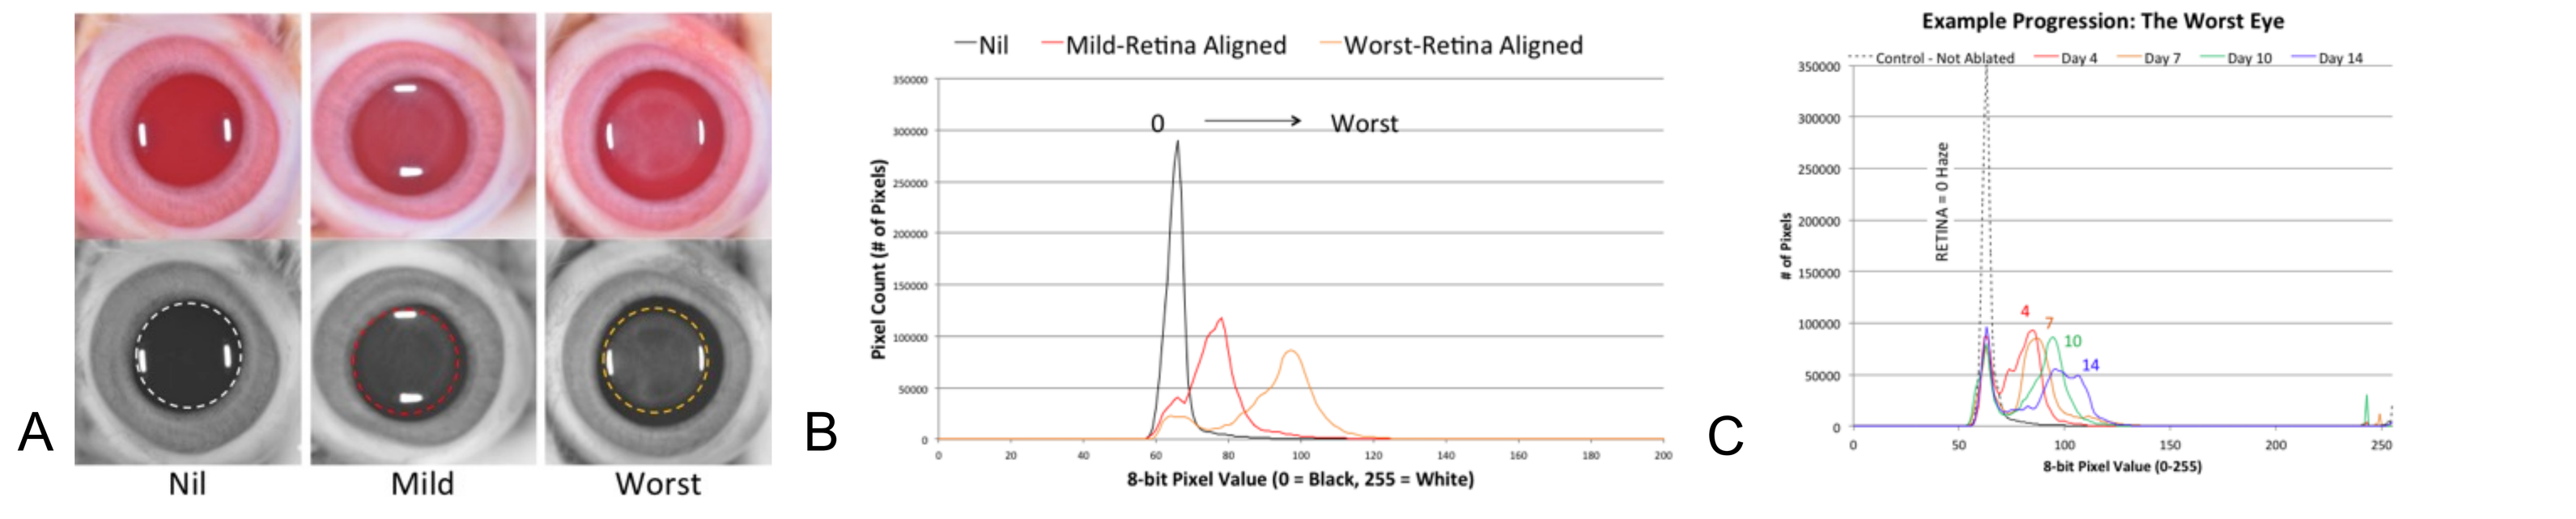
A
Nil
Mild
Worst
B
Nil
Mild-Retina Aligned
Worst-Retina Aligned
0
Worst
Pixel Count (# of Pixels)
8-bit Pixel Value (0 = Black, 255 = White)
C
Example Progression: The Worst Eye
Control - Not Ablated
Day 4
Day 7
Day 10
Day 14
RETINA = 0 Haze
of Pixels
8-bit Pixel Value (0-255)

Figure 2. Haze Histograms. The severity of haze (A) can be measured by the difference between peaks (B, normalized) which are the modes of the retina and haze. C) this can be used to also follow haze progression in a single given eye.

Analysis: A pooled Student's t-Test was used to compare the treatment groups to the control. Additionally, a paired Student's t-Test was used to compare the progress of each eye (paired with its previous measures) over the time period of haze formation (days 4 to 14).

RESULTS

Irritation, Edema, & Re-Epithelialization

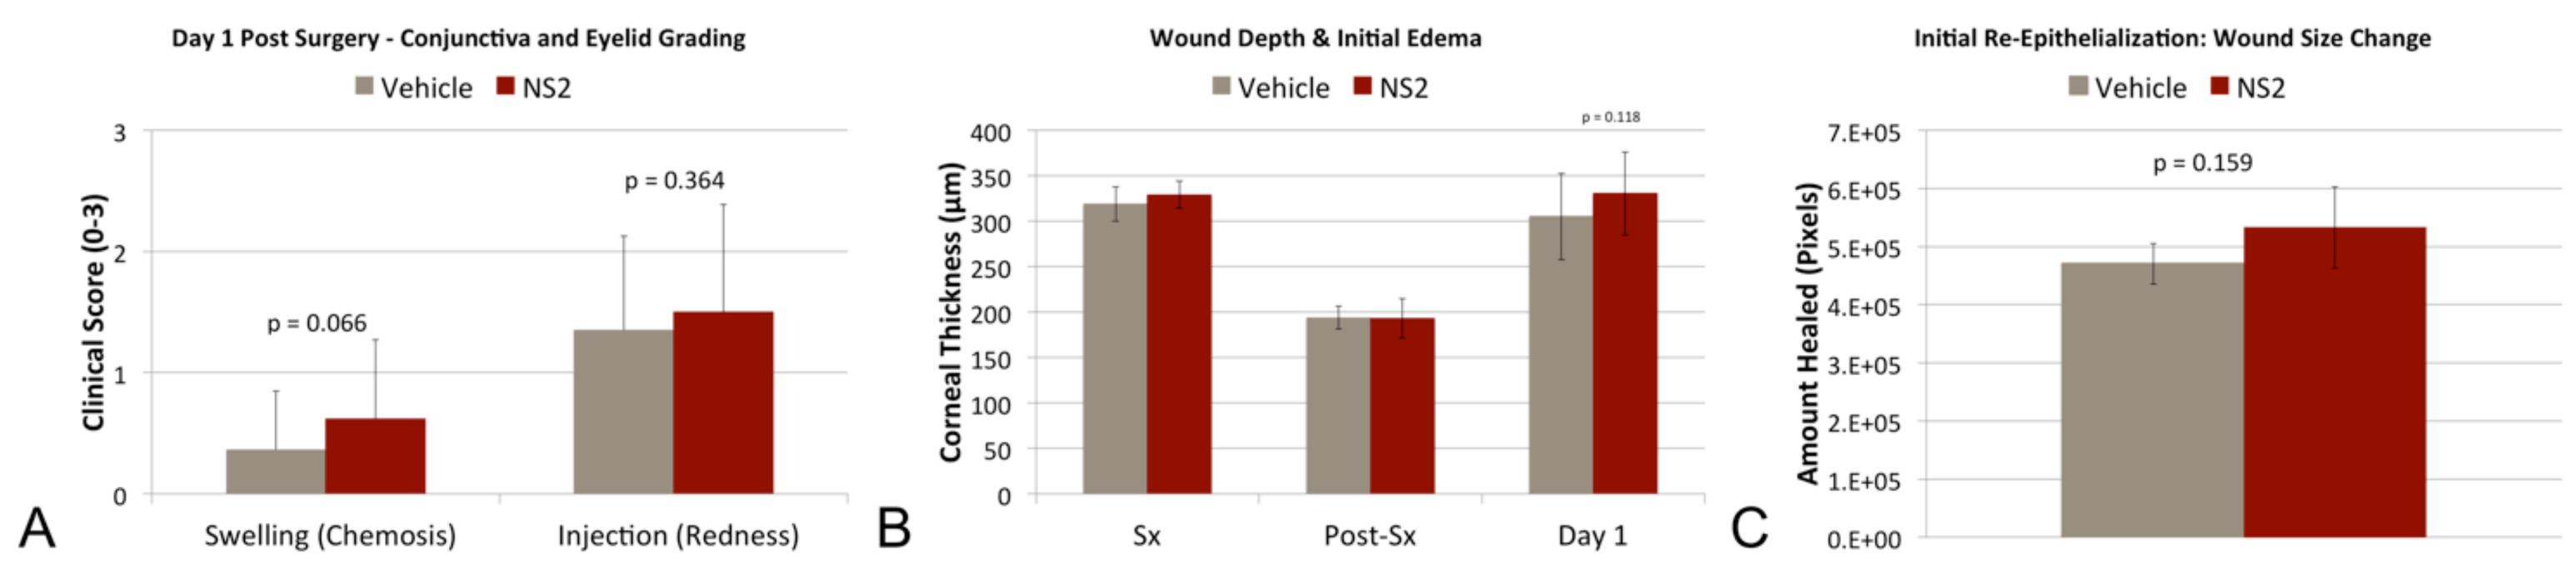
Day 1 Post Surgery - Conjunctiva and Eyelid Grading
Vehicle
NS2
Clinical Score (0-3)
p = 0.066
p = 0.364
Swelling (Chemosis)
Injection (Redness)
A
Wound Depth & Initial Edema
Corneal Thickness (µm)
p = 0.118
Sx
Post-Sx
Day 1
B
Initial Re-Epithelialization: Wound Size Change
Amount Healed (Pixels)
p = 0.159
C

Figure 3: Comparisons of the immediate response to surgery. A) Irritation of the conjunctiva of each eye was clinically graded by 3 individuals using a 0-3 scale for chemosis (swelling) and injection (redness). There was nearly a statistically significant difference, but the overall swelling level was clinically low. B) The corneas' thicknesses were all similar prior to surgery, after surgery, and in the edema present at Day 1 post surgery. C) There was no significant difference in the initial re-epithelialization rate either.

NS2 was not irritating and it did not interfere with wound closure.

Final Corneal Haze

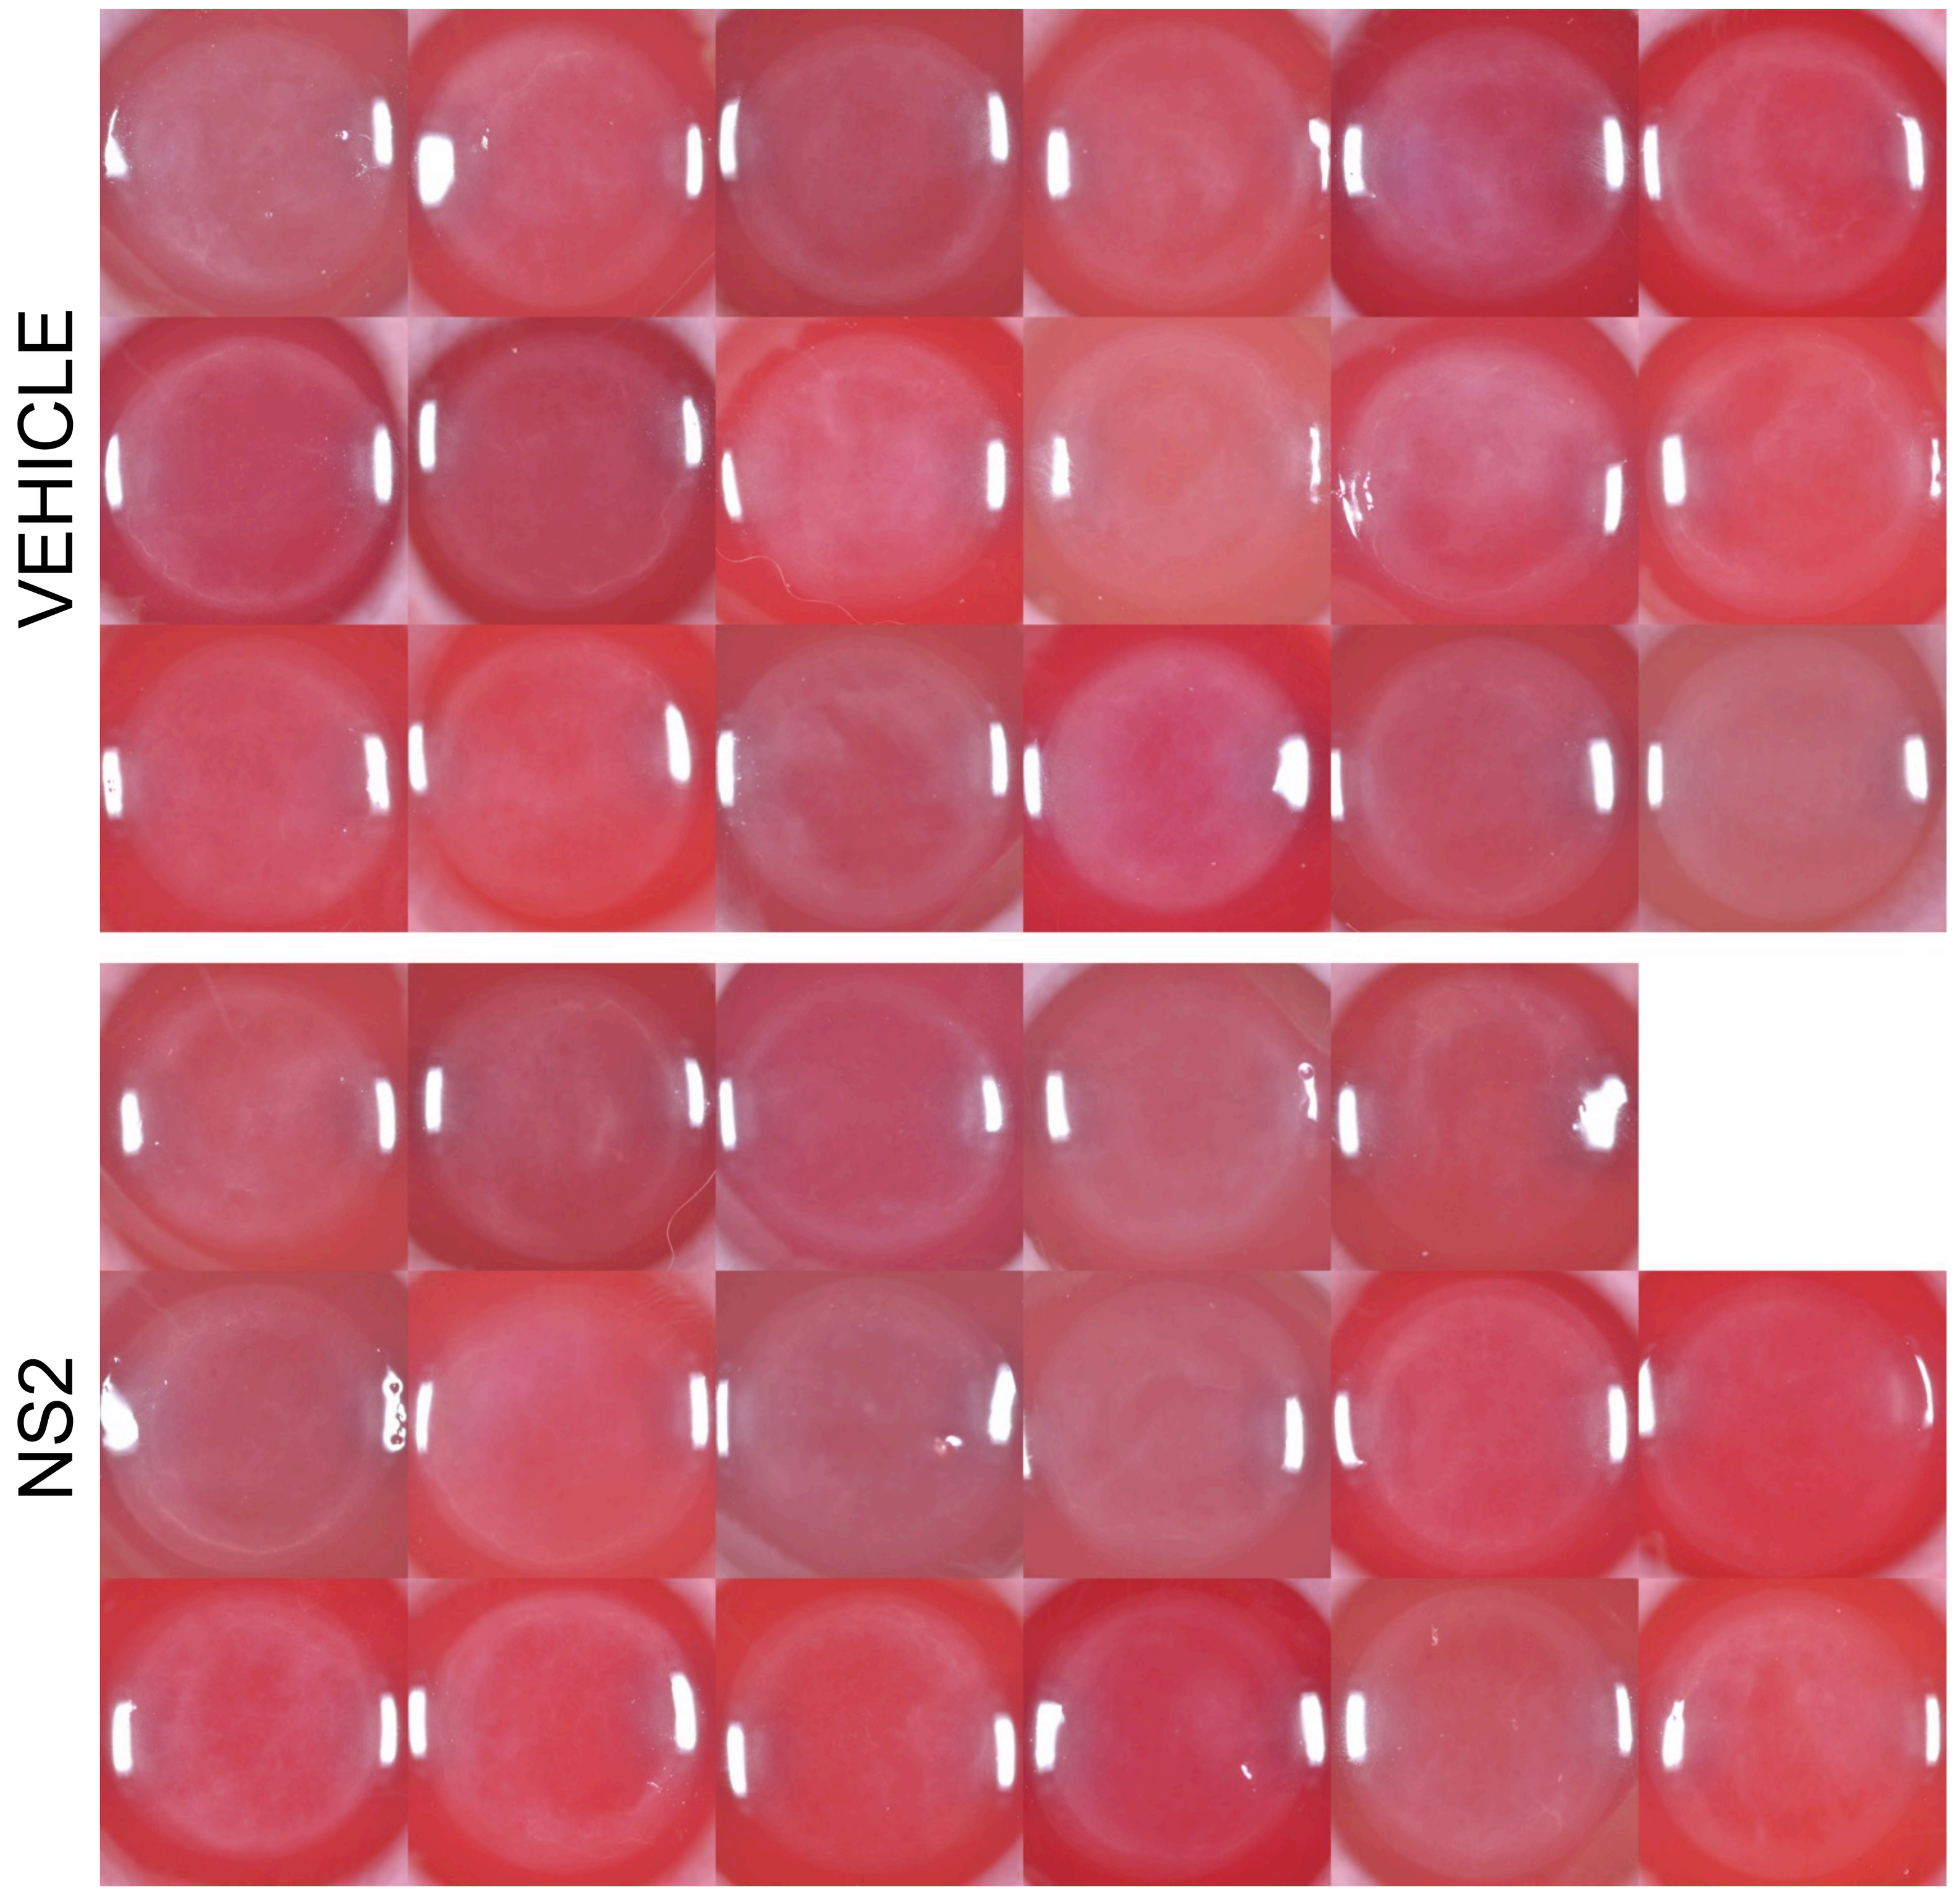
VEHICLE
NS2

Figure 4: Comparisons of the final corneal haze at day 14. Drug-treated corneas still formed haze, but none were as intense as the 3 highest levels in the vehicle group.

RESULTS CONTINUED

Haze Quantification & Progression

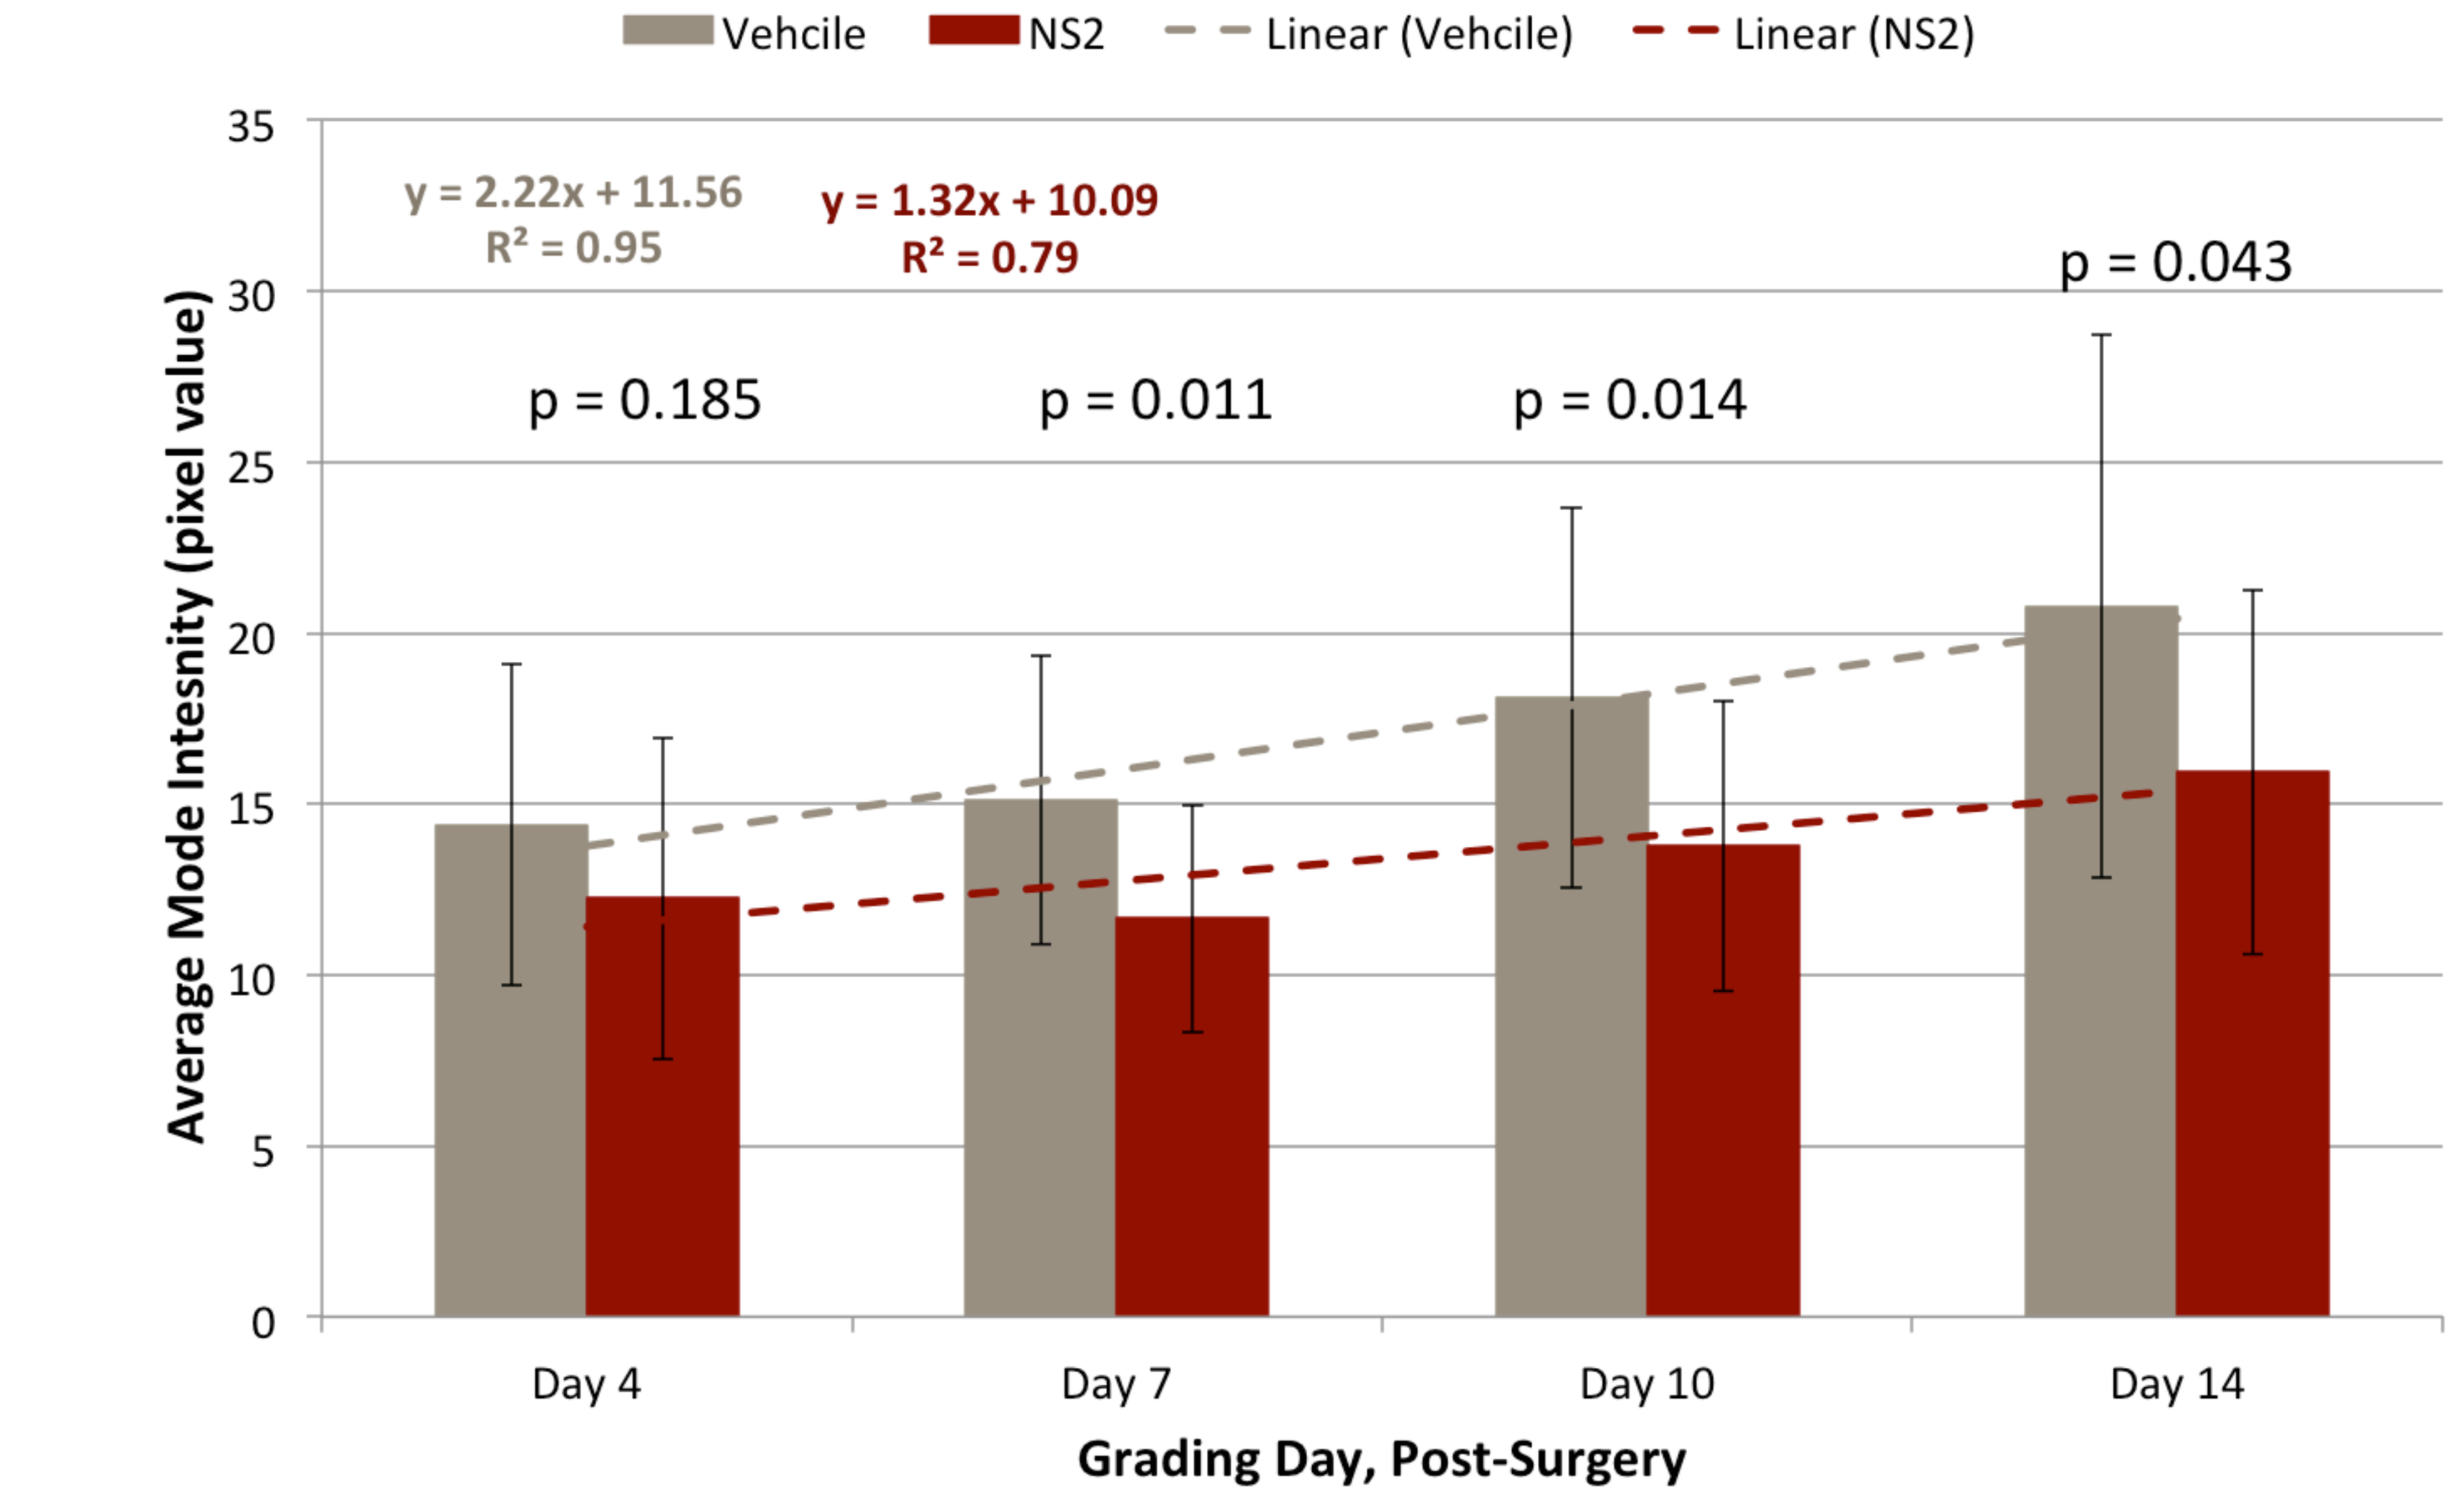
Vehcile
NS2
Linear (Vehcile)
Linear (NS2)
y = 2.22x + 11.56
R² = 0.95
y = 1.32x + 10.09
R² = 0.79
p = 0.185
p = 0.011
p = 0.014
p = 0.043
Average Mode Intesnity (pixel value)
Day 4
Day 7
Day 10
Day 14
Grading Day, Post-Surgery

Figure 5: Differences in corneal haze began to be significant at day 7 post surgery. The drug appears to have reduced the rate of haze formation (slopes) by about 41%.

NS2 lowered the haze intensity and it reduced the rate of the development of haze.

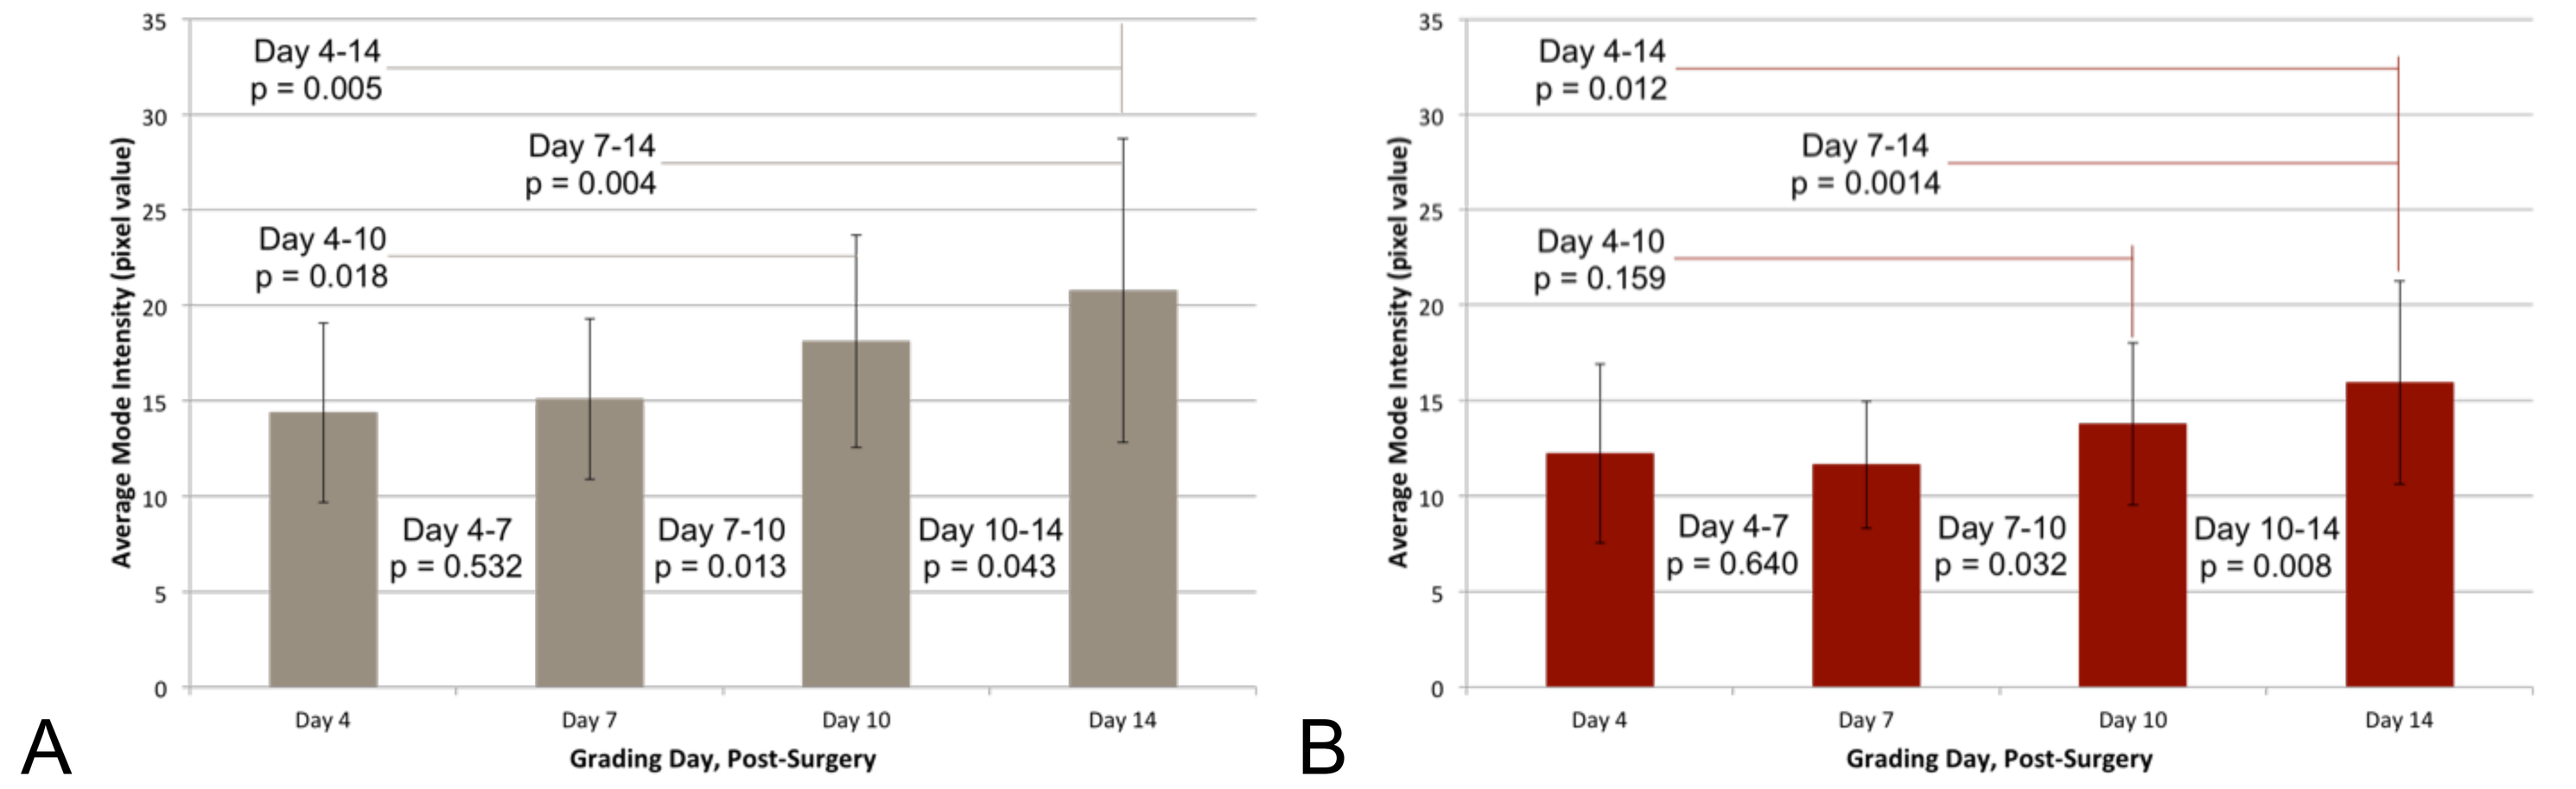
Day 4-14
p = 0.005
Day 7-14
p = 0.004
Day 4-10
p = 0.018
Day 4-7
p = 0.532
Day 7-10
p = 0.013
Day 10-14
p = 0.043
A
Day 4-14
p = 0.012
Day 7-14
p = 0.0014
Day 4-10
p = 0.159
Day 4-7
p = 0.640
Day 7-10
p = 0.032
Day 10-14
p = 0.008
B
Average Mode Intensity (pixel value)
Grading Day, Post-Surgery

Figure 6: Within each group, between each pair of time points, each eye's progression was tested via a paired t-Test for A) the vehicle group and B) the drug group. Neither group had a significant difference until after day 7, while the drug may not have had a significant difference until after day 10 (significant from 4-7, but not 4-10).

The grading method can resolve the progression of haze & NS2 may have delayed the start of the haze formation process.

CONCLUSIONS

The NS2 appears to reduce corneal haze in a rabbit model of PRK. Additional analyses are now being conducted to verify the results biochemically and histologically. The data suggest that the drug has potential to be a novel therapy for PRK and other ocular diseases characterized by inflammation and fibrosis.

Daniel J. Gibson, gibsondj@ufl.edu